Fatigue Reduction and Quality of Life Enhancement Following Therapy with Taurox

Floyd E. Taub, M.D., Char Tara Albert, Lesley Crowder, Suzin Mayerson Wright, Ph.D.
FindCure.Org, Box 2726, Dillon, CO 80435 USA fet@findcure.org 303-249-9174

Objective. Taurox™ has been reported to reduce fatigue in patients with HCV, cancer, Post-Lyme/Chronic Lyme Disease, CFS and Fibromyalgia. Clinical trials demonstrated improvement in QOL measures in addition to a reduction in fatigue. However, clinical trials often report better results than post-marketing studies. We studied QOL benefit in home use, in a generally healthier population not subject to clinical trial selection and the attendant healing environment.

Materials and methods. Taurox (Tauroxicum) is a homeopathic remedy ingredient made from COBAT, a modified di-peptide containing β-alanine and taurine. Taurox is administered in nanogram/day doses sub-lingually. Attempts were made to contact all persons purchasing Taurox products from CureImmune, Inc. If the customer was there, a structured interview was conducted. In order to avoid reply selection bias, no message was left for customers who were not available.

Results. A total of 76 users were reached in three calling periods. Some QOL measures did not change, while other specific measures of QOL were reported to be significantly improved. Fatigue was decreased in over 70% of users; other CNS symptoms (mood and ability to concentrate) and allergy symptoms improved in 40-70% of users.

Conclusion. Users of Taurox report significant improvement in several QOL measures. Neuroimmunologic symptoms, especially fatigue, improved.

Presented at the13th Annual Symposium on Complementary Health Care, Dec 12-14 2006, University of Exeter, UK .
Acknowledgements: FindCure.Org, Inc. and Efficas, Inc. provided funding. This printing contains revisions.

BACKGROUND

Taurox (Molecular Weight: 724 g/m)

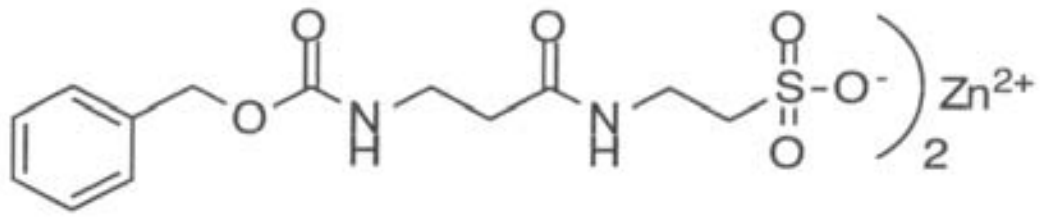

Structure: Taurox™ (Tauroxicum or the Zn salt of Carbo-benzoxy-beta-alanyl taurine [COBAT].

Mouse *in vivo*: This molecule has a variety of immune stimulatory activities and anti-cancer effects (1). Toxicology studies revealed a therapeutic index of ten million (note A).

Human Cells *in vitro*: Taurox enhances components of early T cell activation, including increased intracellular calcium, up-regulation of expression of the CD69 T cell activation marker, enhanced proliferation of peripheral mononuclear cells in culture, and increased granzyme levels (2). Tumor necrosis factor alpha (TNF-alpha) and interferon gamma messenger RNA were up-regulated in quiescent cells and decreased in exogenously stimulated cells (2).

Human Homeopathic Proving: None of 39 normal volunteers reported serious adverse events. In this double blind placebo controlled trial 92% of those receiving Taurox experienced physiologic changes as judged by Dr. David Riley (U.S.) (proving symptoms) while 26% of the placebo group experienced these. (see note B) This difference was statistically significant (Chi-square $p < 0.001$).

Human Clinical Outcome Studies: Physician office based open label clinical outcomes studies were undertaken to evaluate fatigue and other symptoms. The table shows fatigue scores as assessed after 3-6 weeks of Taurox by the FACIT-F (3,4) validated scale. Frequency of allergies and other symptoms were also reported. Further improvement with time was seen.

Placebo-Controlled Double-Blind Proving: Physiologic Impact

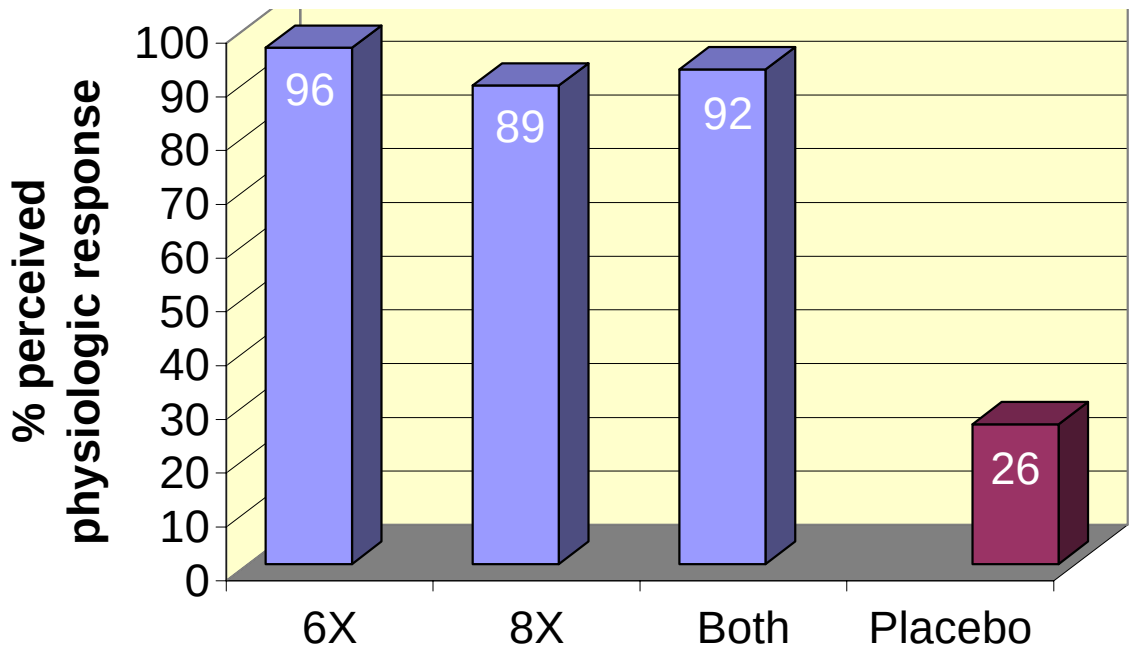
% perceived physiologic response
100
90
80
70
60
50
40
30
20
10
0
96
89
92
26
6X
8X
Both
Placebo

METHODS

A structured interview was conducted with all customers reached who had been taking any Taurox containing product (note C). For each specific symptom, they were asked to categorize their response (columns in the table). Several attempts were made to contact all persons purchasing Taurox products from CureImmune, Inc. In order to avoid reply selection bias, no message was left for customers who were not available. Taurox is administered sublingually in preparations containing from 3X to 30X potencies of Taurox. In total, approximately 15 to 300 nanogram/day of Taurox were received by compliant users. Each preparation contained both molecular and sub molecular potencies of Taurox and of other homeopathic remedies (note C).

As blinding and placebo controls are not possible in post marketing studies internal control questions were included. Users

1

were asked about appetite/eating, which were previously believed to not be affected by Taurox. In addition users were asked about a change in “cravings” (cigarettes, etc.) At the time the study was done, the interviewers thought that people often experienced such a decrease.

Pre-Market Clinical Studies

Indication	N	Mean improvement Of responders	% Improved	Pre- vs Post T-test (2-tailed)
Fatigue (total)	**30**	**49%**	**73%**	**P=0.001**
in HCV	8	51%	87.5%	n/a (small subset of above)
in Cancer	11	55%	73%	n/a (subset)
in CFS	3	37%	100%	n/a (subset)
in Allergies	20	47%	65%	P=0.038 (subset)
Allergies	**20**	**64%**	**60%**	**P=0.027**

RESULTS

A total of 76 customers were reached in three calling periods covering a 1 year period. Some QOL measures did not change. Less than 7% of the people reported a definite decrease in the symptoms of appetite/eating or cravings. In comparison several specific measures of QOL were reported to be significantly improved. Approximately 76% reported an improvement in energy/fatigue. [This includes people reporting that it was either “definitely beneficial” (71%) or “maybe” beneficial (5%)]. Other CNS symptoms, including a “feeling of well being” (74%), ability to concentrate/think (66%) and/or sleep (34%) were also frequently improved. Those people with allergies frequently thought they were or might be experiencing some benefit in the allergic (rhinitis or asthmatic) symptoms (41%). See following table for delineation of symptoms asked about (rows) allowed responses (columns) and percentage of users with each responses. No significant adverse events were noted. Transient headaches, excitement, anxiousness and rarely increased intensity of frequency of heartbeat were reported early in therapy. In some cases this led to usually transient dose reduction. One person reported being over-energized and not sleeping as well, he typically took the product late in the day (contrary to the instructions). See graph last page.

Abbreviations: CFS – chronic fatigue syndrome, CNS – central nervous system; EPO – erythropoietin; QOL – quality of life; HCV – hepatitis C virus.

Structured Interview Responses (Percentages)

	Definitely beneficial	Maybe beneficial	No Effect	Maybe made worse	Definitely made worse	Don’t know; not applicable
Cravings	6.6	3.9	47.4	0	0	42.1
Eating/ Appetite	6.6	7.9	63.2	0	0	22.4
Use Stimulants	10.5	9.2	48.7	0	0	31.6
Pain	15.8	17.1	30.3	0	0	36.8
Sleep Quality	23.7	10.5	50.0	2.6	0	13.2
Allergies/ Asthma	23.7	17.1	22.4	0	0	36.8
Concentrate/ Think	48.7	17.1	19.7	0	0	14.5
Well Being	53.9	19.7	18.4	0	0	7.9
Energy/ Fatigue	71.1	5.3	18.4	0	0	5.3

Table shows percent of total, n=76.

DISCUSSION

Fatigue is the most frequent complaint in most medical practices; 30-50% of the U.S. population experiences fatigue (5). Fatigue significantly decreases QOL in most all persons with chronic disease. Treatment of anaemia may relieve fatigue however treatment with erythropoietin (EPO), may accelerate the growth of some cancers and have potentially fatal side effects (6). EPO gives on average a less than 5 point overall improvement in the FACIT-F (0-53 point) fatigue score (7) while Taurox provided on average a 12 point improvement in the score in pre market clinical studies (described above). An improvement of 3-5 points is considered to be clinically meaningful (4, 7, 8).

The clinical trial healing environment and patient selection bias often lead to greater apparent therapeutic benefit in clinical trials than is observed in the subsequent post marketing use of a drug. However Taurox benefit occurs independent of physician influence. A number of other facts also suggest that the reduction in fatigue seen is not due to placebo or bias. Based on previous anecdotal reports, the interviewers believed there would be a change in cravings. Both the user and interviewer bias would be to expect a

2

decrease in stimulant use, however little benefit was reported in these areas. In studies of fatigue, reduction by EPO injections were not sufficient to cause a decrease in fatigue, only subjects with the largest increase in Hg reported fatigue reduction (8). The CFS literature indicates that placebo effect (and the effect of most tested therapies) is between 0 and 25% (9). Thus both internal test questions and the scientific literature suggest that the level of fatigue reduction in this study cannot be accounted for by placebo effect.

CONCLUSIONS

Taurox produces similar beneficial effects in the home environment, as in studies associated with the physician office healing environment. Taurox improves several CNS related symptoms. Fatigue was the most frequently improved symptom. Participants also reported an improved “feeling of well being” and improved “concentration/ability to think”.

References:

1. Knight, G. D., Laubscher, K. H., Fore, M. L., Clark, Scanlen, T. J. (1994). Vitalethine Modulates Erythropoiesis and Neoplasia. Cancer Research, 54(21): 5623-35.
2. Dunn T, Taub F, Pontzer C, *Immunostimulatory Effects of Taurox SB™*, U MD, Dept. of Cell Biology and Molecular Genetics, manuscript submitted for publication.
3. Cella, D. (1997). The Functional Assessment of Chronic Illness Therapy (FACIT) Measurement System Manual. Evanston Northwestern Healthcare, Evanston, IL USA.
4. Kimberly Webster, K., Cella, D. and Yost, K. (2003) The Functional Assessment of Chronic Illness Therapy (FACIT) Measurement System: properties, applications, and interpretation. Health and Quality of Life Outcomes, 1:79.
5. Jason, L.A. (2003) AACFS 6th International Conference, January 31, 2003 (www.co-cure.org/aacfs6.htm).
6. EPO package insert includes warnings concerning increased incidence of cardiac arrest, neurologic events (including seizures and stroke), exacerbations of hypertension, congestive heart failure, vascular thrombosis/ischemia/infarction, acute myocardial infarction, and fluid overload/edema. These increase with increased response to EPO.
7. Berndt, E., Kallich, J., McDermott, A., Xu, X., Lee, H. and Glaspy J. (2005) Reductions in Anaemia and Fatigue are Associated with Improvements in Productivity in Cancer Patients Receiving Chemotherapy. J. Pharmacoeconomics, 23(5): 505-514.
8. Cella, D, Lai, J.S., Chang, C.H., Peterman, A., & Slavin, M. (2002). Fatigue in cancer patients compared with fatigue in the general population. Cancer, 94(2), 528-538.
9. Carette S, Bell MJ, Reynolds WJ, Haraoui B, McCain GA, Bykerk VP, Edworthy SM, Baron M, Koehler BE, Fam AG, et al. (1994). Comparison of amitriptyline, cyclobenzaprine, and placebo in the treatment of fibromyalgia: A randomized, double-blind clinical trial. Arthritis Rheum, 37:32-40.
10. Hannonen P, Malminiemi K, Yli-Kerttula U, Isomeri R, Roponen P. (1998). A randomized, double-blind, placebo-controlled study of moclobemide and amitriptyline in the treatment of fibromyalgia in females without psychiatric disorder. Br J Rheumatol. Dec;37(12):1279-86.
11. Arnold LM, McElroy SL, Hudson JI, Welge JA, Bennett AJ, Keck PE. (2002). A placebo-controlled, randomized trial of fluoxetine in the treatment of binge-eating disorder. J Clin Psychiatry. Nov; 63(11):1028-33.

% Reporting QOL Benefits

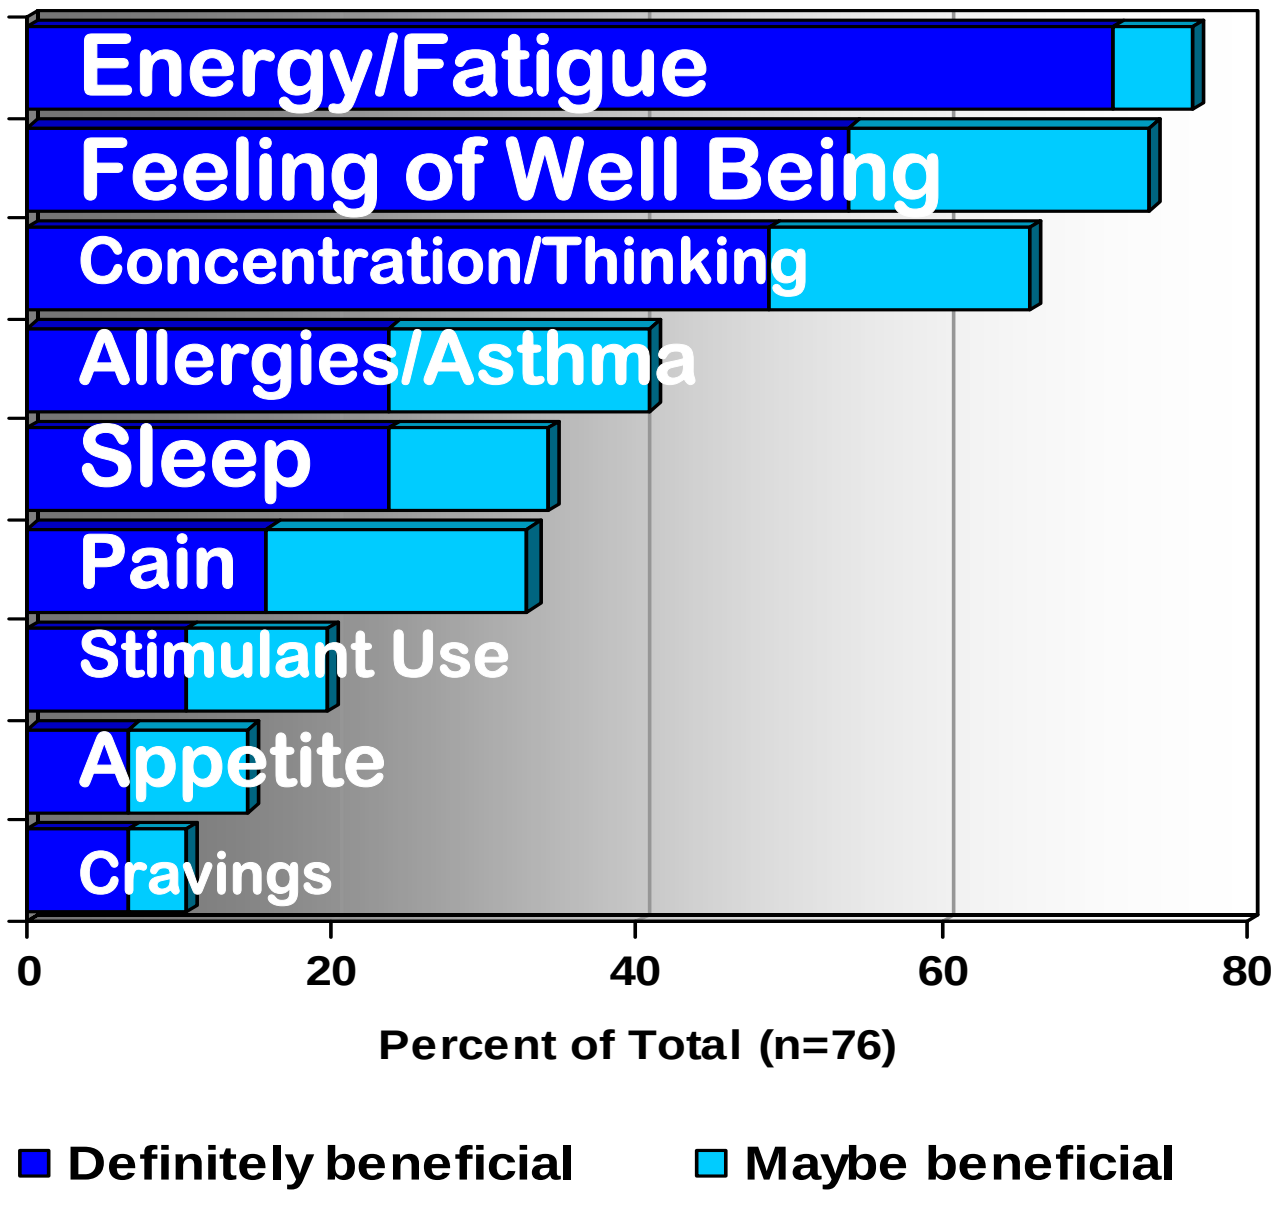

Energy/Fatigue
Feeling of Well Being
Concentration/Thinking
Allergies/Asthma
Sleep
Pain
Stimulant Use
Appetite
Cravings
0
20
40
60
80
Percent of Total (n=76)
Definitely beneficial
Maybe beneficial

Notes:

A. Safety of Taurox was assessed in 4 rodent studies, 2 dosed by IV injection (single dose studies), and 2 dosed by oral gavage (multiple dose studies). Taurox did not produce any gross signs of toxicity when administered by oral gavage to rats for 14 days at a dose of 2,000 mg/kg/day. Due to the low dose required for effectiveness in humans, this represents a safety factor > ten million or 10^7.

B Details of the proving and the Materia Medica will be presented separately.

C. Products. More details available on the web site. Percentage of sample population using each product is indicated. Some used more than one product.

Taurox with minerals, 7X to 30X. Users purchased this product primarily for cold, flu, allergies and mild or moderate symptoms. (19% of customers).
Taurox with herbs, 6X to 30X. Users purchased this product primarily for fatigue and more severe symptoms. (49% of customers).
Taurox with no other ingredients, 3X, 6X and 9X, primarily for fatigue and other symptoms associated with various medical conditions. (38% of customers) This is equivalent to a 5X dose, molecularly.

3